Characterization of side chain michael acceptors based tissue transglutaminase inhibitors

Ralf Pasternack[1], Kai Oertl[1], Johannes Weber[1], Ina Lindemann[2], Gerhard Klebe[2] and Martin Hils[1]
[1]Zedira, Darmstadt, Germany and [2]Philipps University Marburg, Marburg, Germany

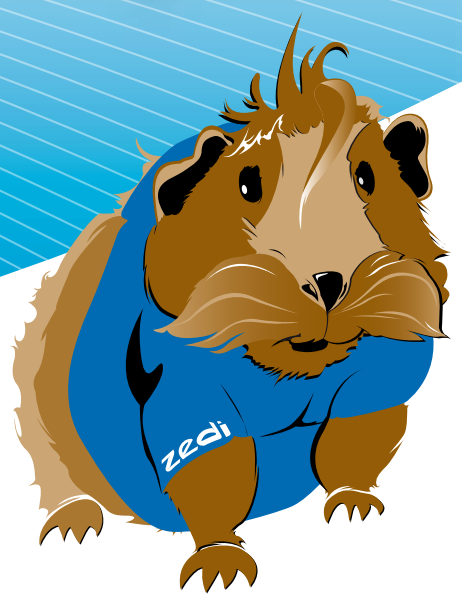

Introduction

Tissue Transglutaminase (TG2) is ubiquitously expressed throughout the human body. Obviously, TG2 expression and activation is a key function in inflammatory events. Further, TG2 is involved in the pathogenesis of celiac disease. The interaction between gliadin and potentially dysregulated transglutaminase activity might induce a vicious circle of inflammation and deamidation. We are interested in the discovery of potent and selective TG2 inhibitors to intervene in the pathologic event.

Approach

Based on peptide screening we identified the tetrapeptide Z-Gln-Gln-Pro-Leu-OMe being an excellent substrate for tissue transglutaminase using a transamidation read out. The search for suitable warheads led us to the well known michael acceptors targeting thiol dependent enzymes.

Chemistry

To introduce the phamacophore, the reactive glutamine had to be replaced by the michael acceptor. We developed a multi step synthesis to yield a building block compatible with solid phase chemistry. The ZED754 lead compound was used to disclose the molecular interaction with tissue transglutaminase‘s active site.

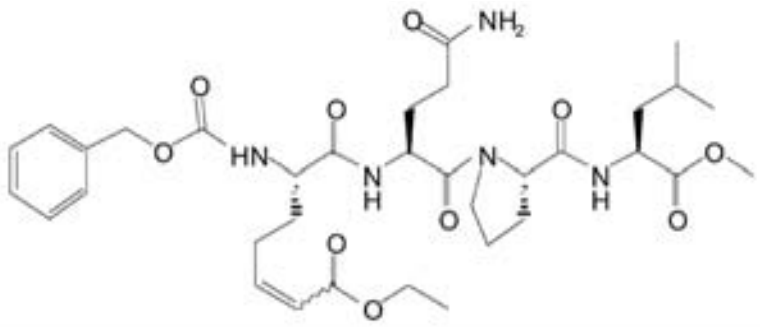

ZED754
peptidic lead compound containing the thiol reactive michael acceptor warhead.

Crystallization and x-ray structure

The crystal structure of TG2 in complex with the covalently bound peptidic lead compound ZED754 was determined at 2.5 Å resolution. Conditions for crystallization were similar to those described by Pinkas, D.M. et al. (2007) PLoS Biology. We could confirm the same crystal morphology and conformation of the peptide backbone in the primary binding site.

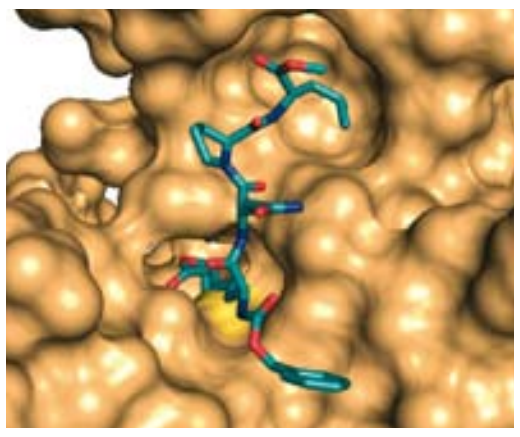

The inhibitor (shown in cyan) is covalently bound to the active site cysteine located in the catalytic tunnel. Tight binding molecules have to address the hydrophobic pocket in addition. The proline residue obviously directs the corresponding ligand (leucine) to fit into this pocket.

Medicinal chemistry

To design druggable compounds the “QPL-OMe” conformational motive was replaced using peptidomimetics. The resulting scaffold was used to learn more about structure activity relations (SAR).
200 analogues were prepared optimizing the potency and selectivity. The best compounds ZED1098, ZED1219 and ZED1227 were profiled in detail against different human transglutaminases and for stability in body fluids. Further, promising data concerning cytotoxicity and acute toxicity in rats were obtained. The candidates will be further assessed in advanced preclinics.

Compound	**1227**	**1098**	**1219**
Inhibitory in vitro efficacy (IC_{50}) tissue transglutaminase (TG2)	45 nM	32 nM	25 nM
Selectivity (IC_{50}TGx/IC_{50}TG2)			
FXIIIa	410	230	800
TG1	240	200	195
TG3	340	175	224
TG6	145	75	55
Stability in artificial fluids			
Gastric juice (24 h, 37°C)	stable (95%)	stable (98%)	insoluble at low pH
Intestinal juice (24 h, 37°C)	stable (82%)	stable (83%)	stable (94%)

Financial support from the German Ministry for Education and Research (BMBF) is gratefully acknowledged.

Zedira GmbH, Roesslerstrasse 83, 64293 Darmstadt, Germany **Phone:** + 49 61 51 / 32 51 00 **Fax:** + 49 61 51 / 32 51 19 **E-mail:** contact@zedira.com **Web:** www.zedia.com